BRIGHT MINDS

Bright Minds Biosciences Inc. New York, NY, USA
Bright Minds Biosciences Pty Ltd, Carlton South VIC, Australia

BMB-101 and Biased 5-HT2C Agonism: A Novel Approach for Sustained Epilepsy Management

A Vasilkevich[1], A Lovera, J Duan[1], I McDonald[1], J Sourbron[2], SD Collins[1], J McCorvy[3], JT Pedersen[1]

[1]Bright Minds Biosciences Inc. New York, NY, USA; Bright Minds Biosciences Pty Ltd, Carlton South VIC, Australia; [2]Ghent University Hospital, Ghent, Belgium; [3]Medical College of Wisconsin, Milwaukee WI

2025 AES ANNUAL MEETING
DECEMBER 5-9 • ATLANTA

Background

- **The 5-HT$_{2C}$ receptor (serotonin 2C receptor), a G protein-coupled receptor (GPCR), emerged as a potential target for epilepsy treatment due to its role in modulating neurotransmitter release and neuronal excitability**
- **Traditional 5-HT$_{2C}$ agonists can lead to receptor desensitization, through receptor internalization, due to β-arrestin recruitment, reducing long-term efficacy[1]**
- **Lorcaserin was found to highly desensitize 5-HT$_2$ receptors, more than 5-HT, possibly due to a "super agonist" action at recruiting β-arrestin[2]**
- **Reduced fenfluramine and lorcaserin long-term efficacy as anti-obesity drugs has been proposed to be caused by β-arrestin recruitment, leading to removal of receptors from the cell surface leading to tolerance[3]**

Lessons from lorcaserin

- Deactivation of β-arrestin, using a selective β-arrestin inhibitor, produces a superior and sustained effect in long-term lorcaserin use (in vivo study)

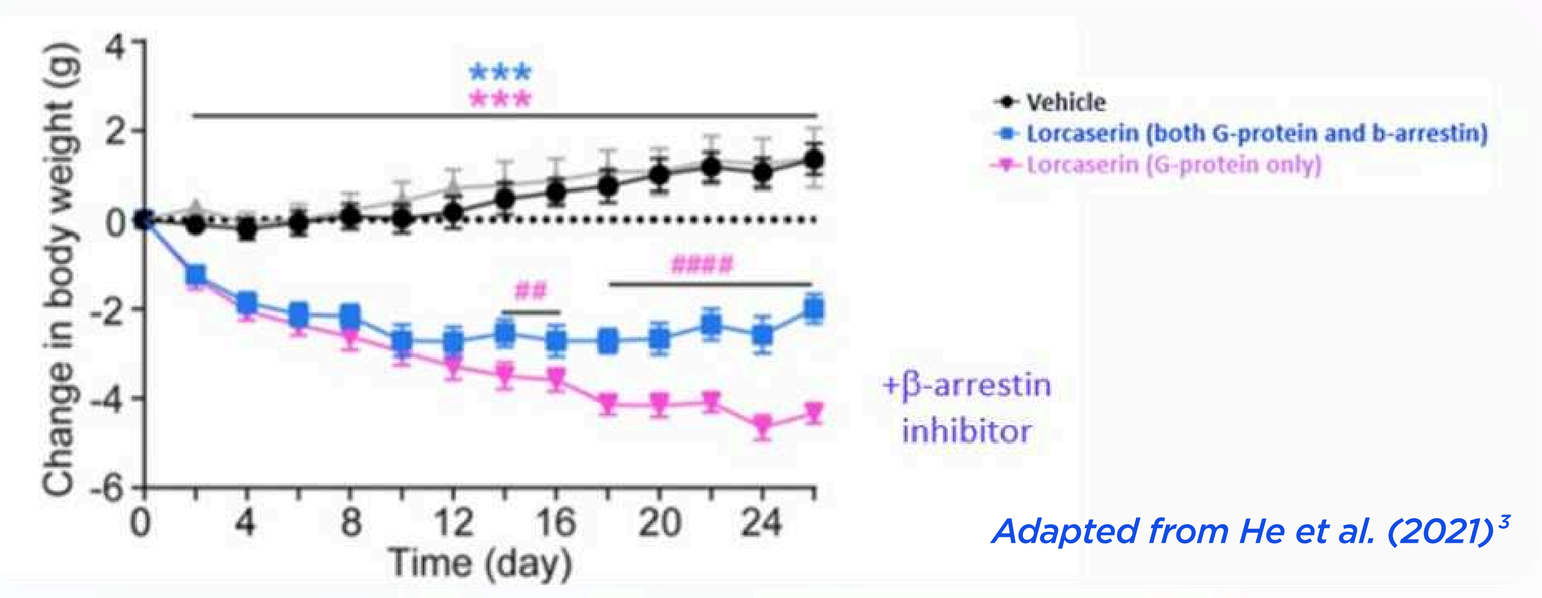
Change in body weight (g)
Time (day)
Vehicle
Lorcaserin (both G-protein and b-arrestin)
Lorcaserin (G-protein only)
+β-arrestin inhibitor

Adapted from He et al. (2021)[3]

BMB-101 is a highly selective 5-HT$_{2C}$ receptor agonist and does not recruit β-arrestin

- In vitro pharmacology (J. McCorvy group): Activation of 5 HT receptors was measured using Gq dissociation as measured by bioluminescence resonance energy transfer (BRET). Emax was defined relative to serotonin (5-HT).
- BMB-101 is a full 5-HT$_{2C}$ agonist with minimal activity at 5-HT$_{2A}$ and 5-HT$_{2B}$ receptors, thus not possessing liabilities linked to psychedelic and cardiotoxic effects, respectively
- BMB-101 shows a great preference for Gq dissociation at 5-HT$_{2C}$ vs. β-arrestin 2 (β-arr2) recruitment (β-arr2: Emax <20%, EC$_{50}$= not determinable), in contrast to norfenfluramine and lorcaserin which are balanced agonists recruiting both Gq and β-arrestin

Compound	5-HT$_{2A}$	5-HT$_{2B}$	5-HT$_{2C}$
BMB-101	2280	>10000	16.2
Norfenfluramine	82.8	11.6	2.5
Lorcaserin	50.1	67.4	2.4
Bexicaserin	>10000	>10000	15 (42*)

Internal BRET data for BMB-101, norfenfluramine, lorcaserin and bexicaserin.
**Value reported by Ren et al (2025)[4]*

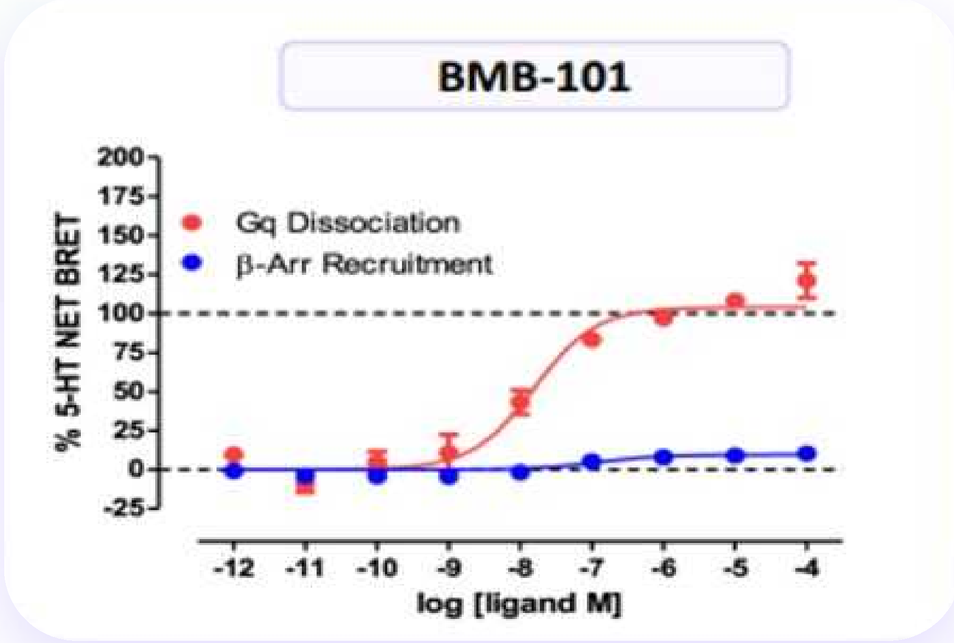
BMB-101
Gq Dissociation
β-Arr Recruitment
% 5-HT NET BRET
log [ligand M]

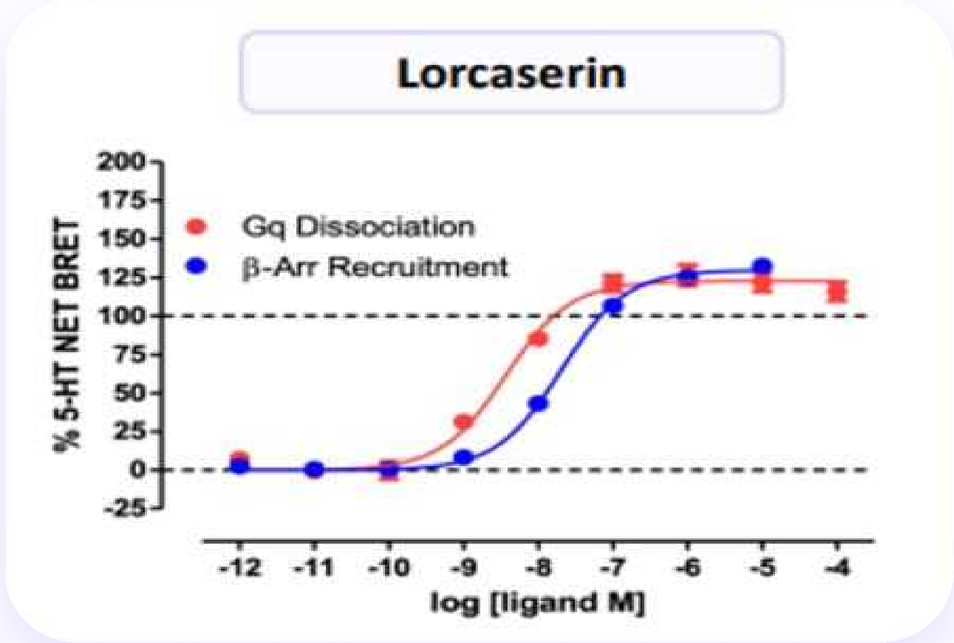
Lorcaserin
Gq Dissociation
β-Arr Recruitment
% 5-HT NET BRET
log [ligand M]

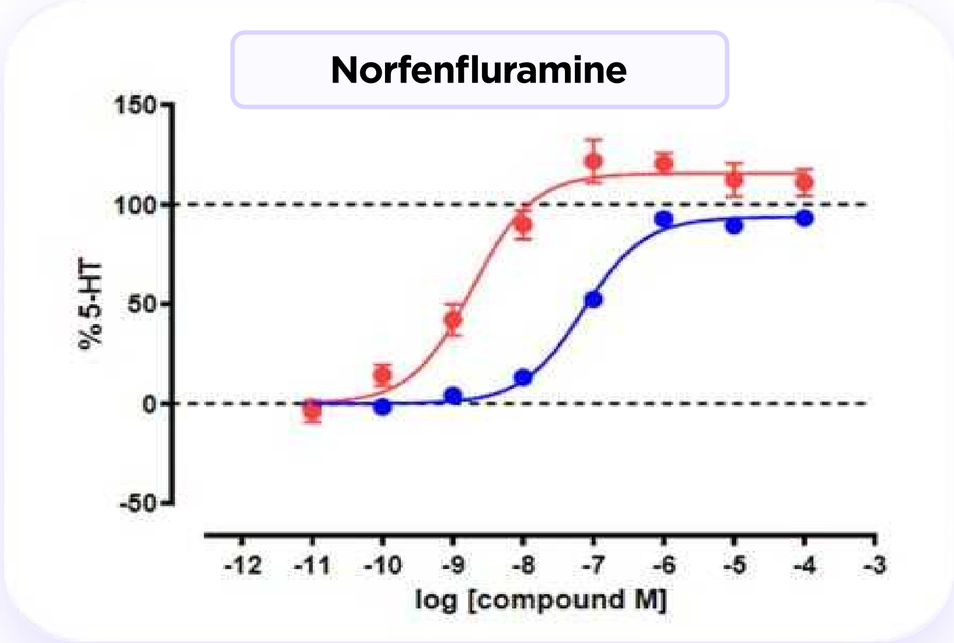
Norfenfluramine
% 5-HT
log [compound M]

Biased agonism: BMB-101 does not recruit any β-arrestin subtype

- BMB-101 was subsequently tested for recruitment of β-arrestin subtypes (β-arrestin 1 [β arr1] and β-arr2) and compared to lorcaserin
- BMB-101 exhibits very weak partial agonism of β-arr1 and β-arr2 recruitment (β-arr1 Emax 12.9%; β-arr2 Emax 20.8%) compared to Gq dissociation
- In contrast, lorcaserin is a superagonist at β-arr1 and β-arr2 (β-arr1Emax 100.3%; β-arr2 Emax 107.4%)
- BMB-101 again shows a greater preference for Gq dissociation agonist activity compared with lorcaserin, demonstrating Gq-biased agonism effects robust across time (15, 60, 120, and 300 minutes)

BMB-101 5-HT$_{2C}$-Gq

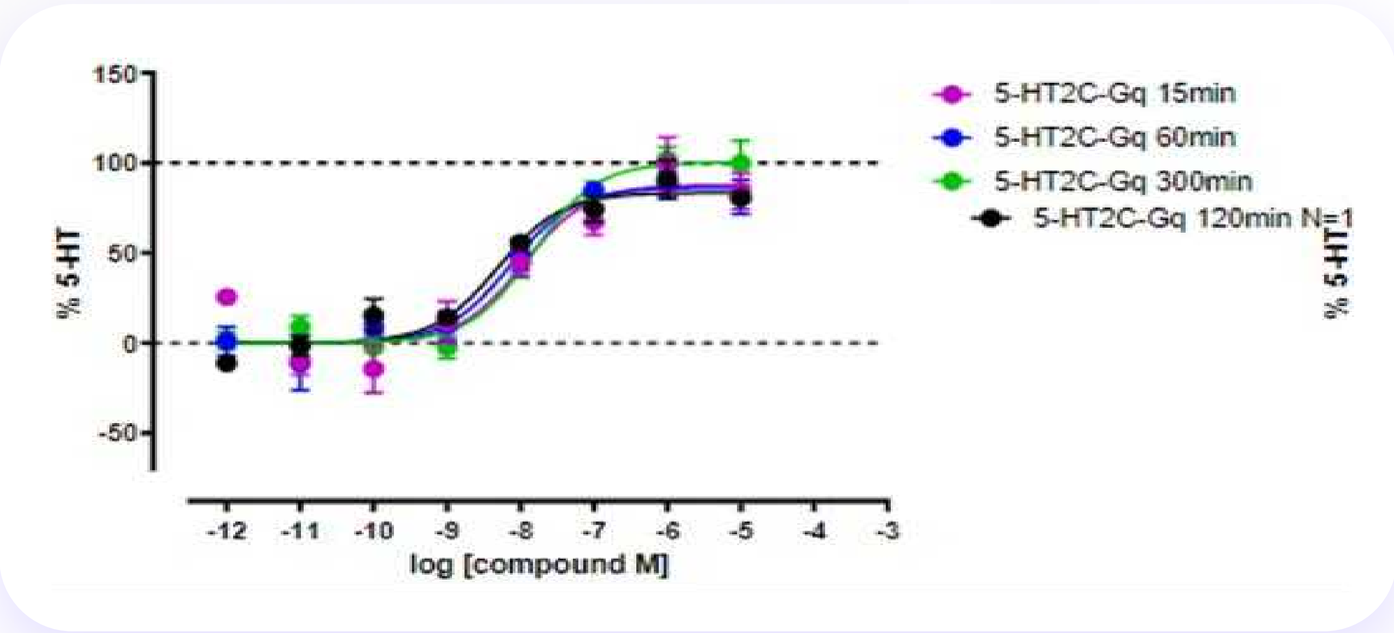
5-HT2C-Gq 15min
5-HT2C-Gq 60min
5-HT2C-Gq 300min
5-HT2C-Gq 120min N=1
log [compound M]

Lorcaserin 5-HT$_{2C}$-Gq

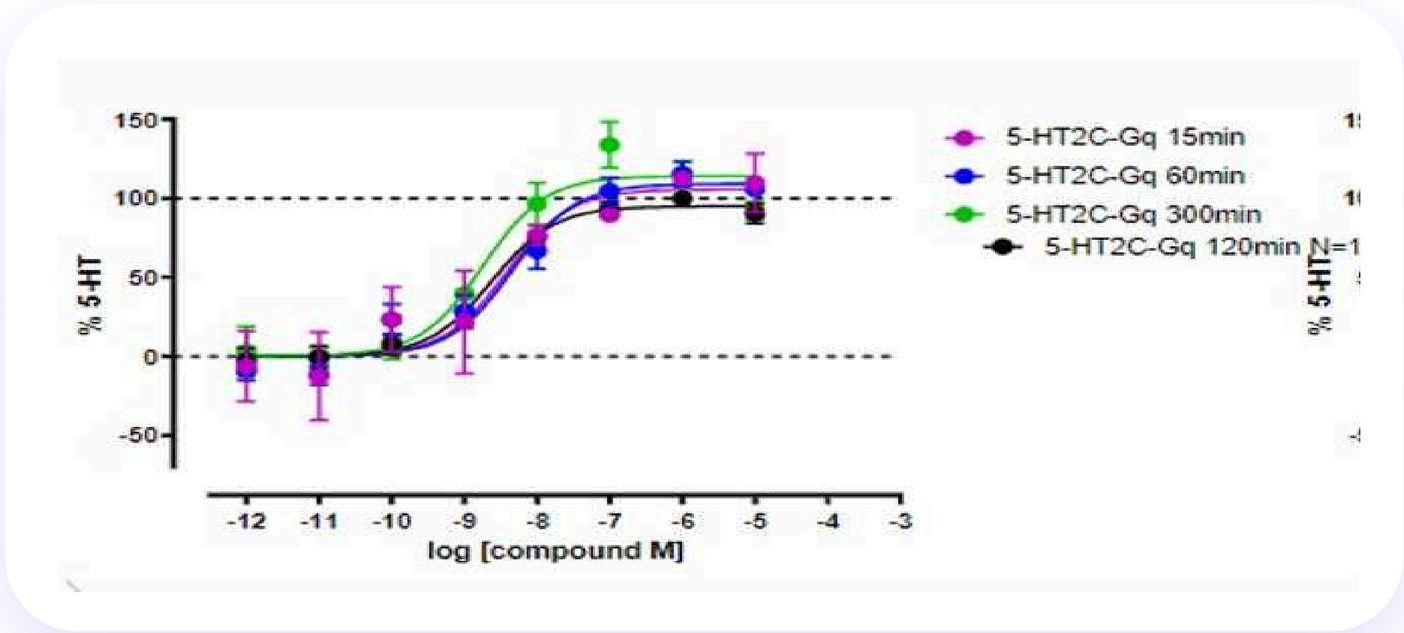
5-HT2C-Gq 15min
5-HT2C-Gq 60min
5-HT2C-Gq 300min
5-HT2C-Gq 120min N=1
log [compound M]

BMB-101 5-HT$_{2C}$-β-arr1

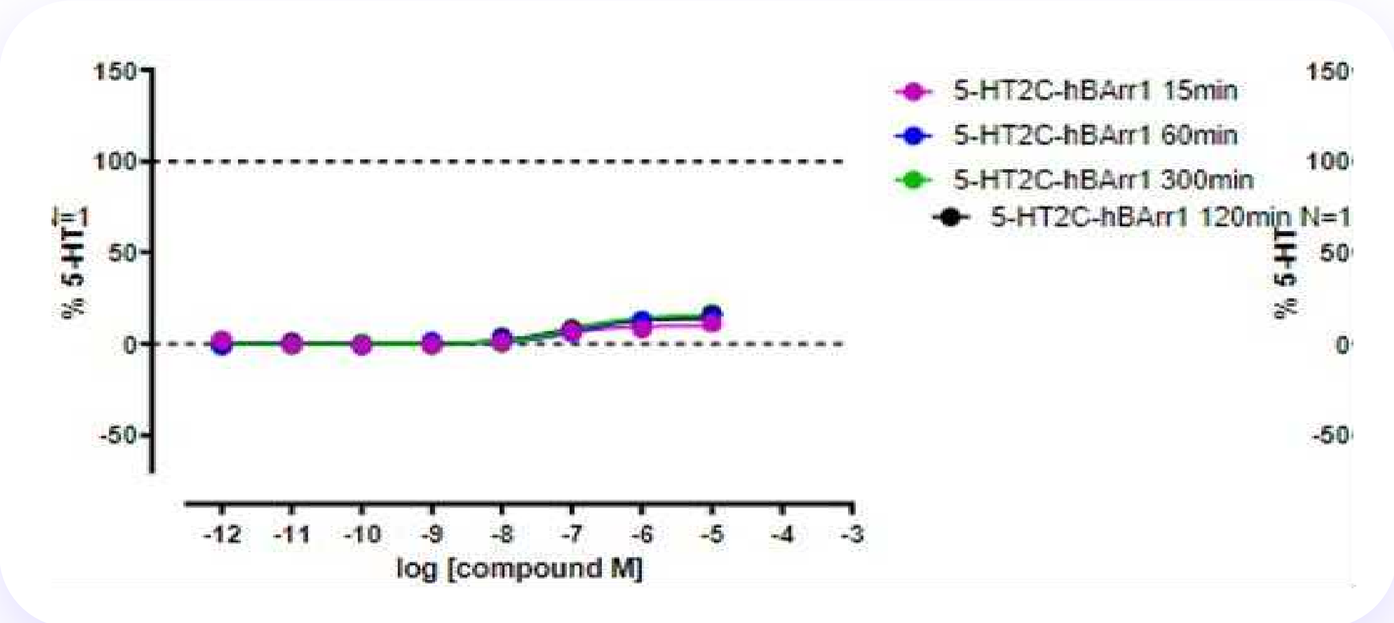
5-HT2C-hBArr1 15min
5-HT2C-hBArr1 60min
5-HT2C-hBArr1 300min
5-HT2C-hBArr1 120min N=1
log [compound M]

Lorcaserin 5-HT$_{2C}$-β-arr1

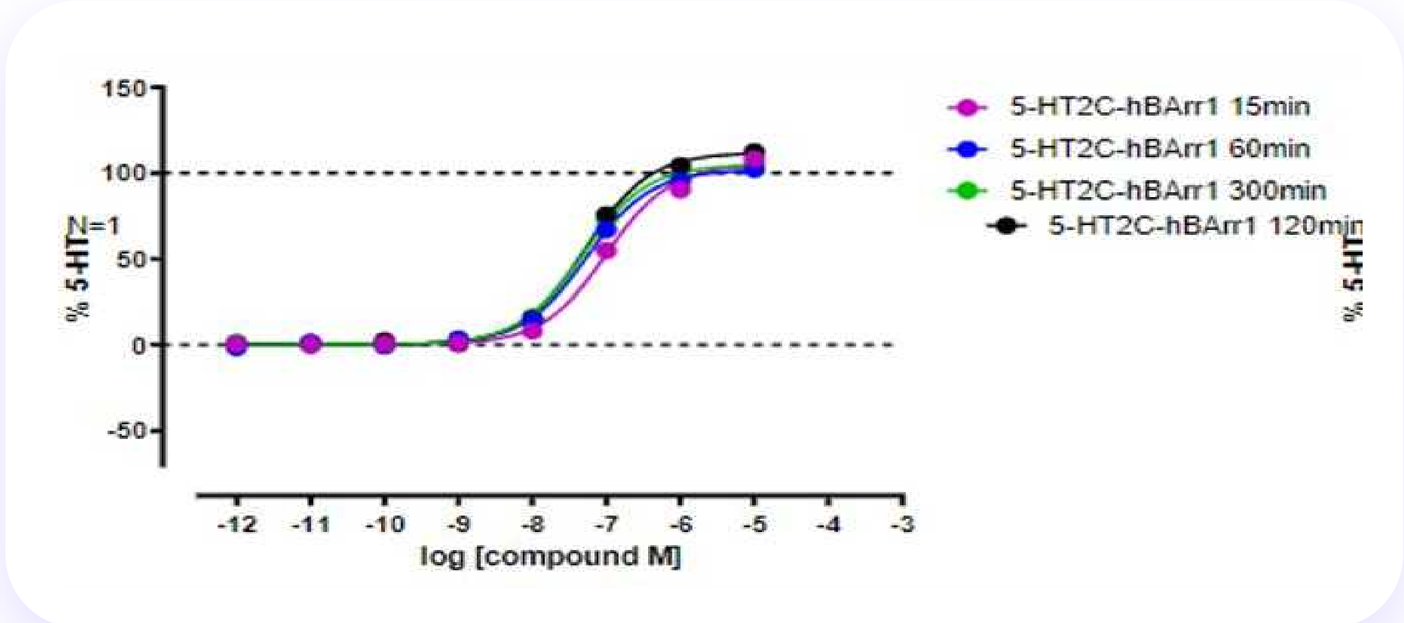
5-HT2C-hBArr1 15min
5-HT2C-hBArr1 60min
5-HT2C-hBArr1 300min
5-HT2C-hBArr1 120min
log [compound M]

BMB-101 5-HT$_{2C}$-β-arr2

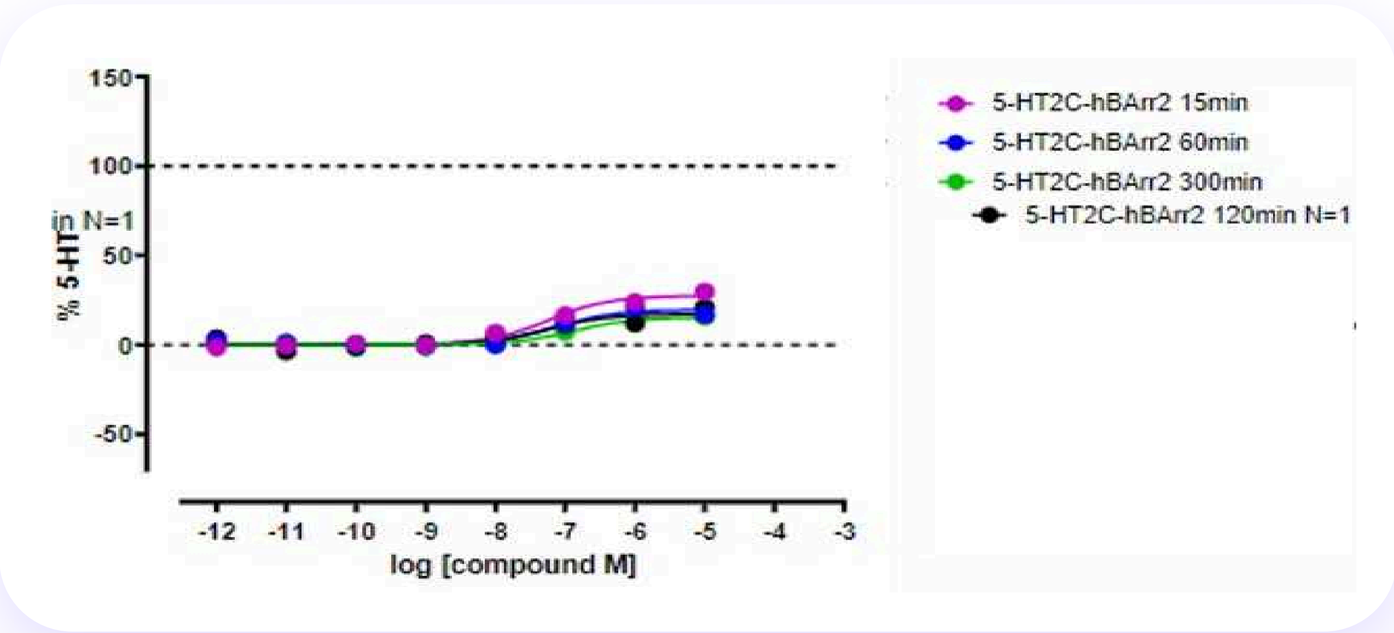
5-HT2C-hBArr2 15min
5-HT2C-hBArr2 60min
5-HT2C-hBArr2 300min
5-HT2C-hBArr2 120min N=1
log [compound M]

Lorcaserin 5-HT$_{2C}$-β-arr2

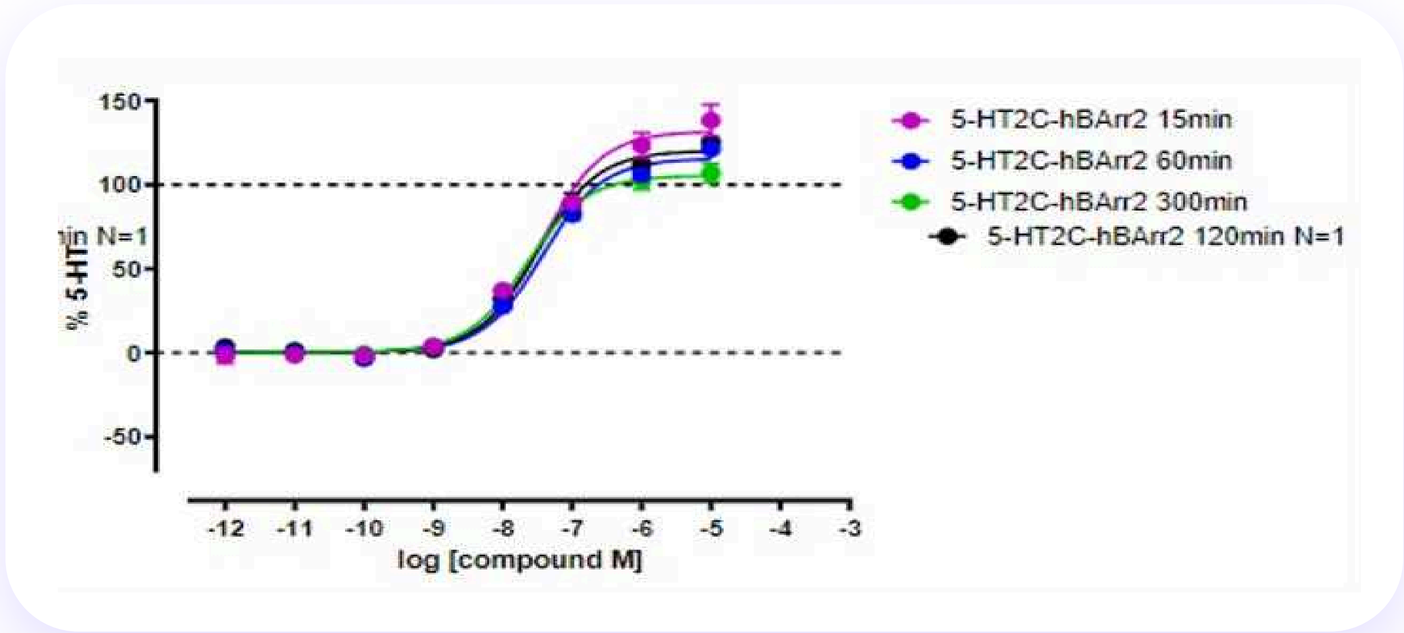
5-HT2C-hBArr2 15min
5-HT2C-hBArr2 60min
5-HT2C-hBArr2 300min
5-HT2C-hBArr2 120min N=1
log [compound M]

Biased agonists: in vivo efficacy

scn1Lab zebrafish model (Dravet Syndrome model)

- Epileptiform activity as duration and frequency of epileptiform events for scn1Lab-/- treated larvae with BMB-101
- Statistically defined outliers were removed
- BMB-101 reduced locomotor activity and reduced the cumulative duration of epileptiform events in the brain
- Hence, the data provide clear evidence that BMB-101 has a therapeutic effect on the abnormal brain activity of scn1Lab zebrafish resulting in a pronounced antiseizure activity

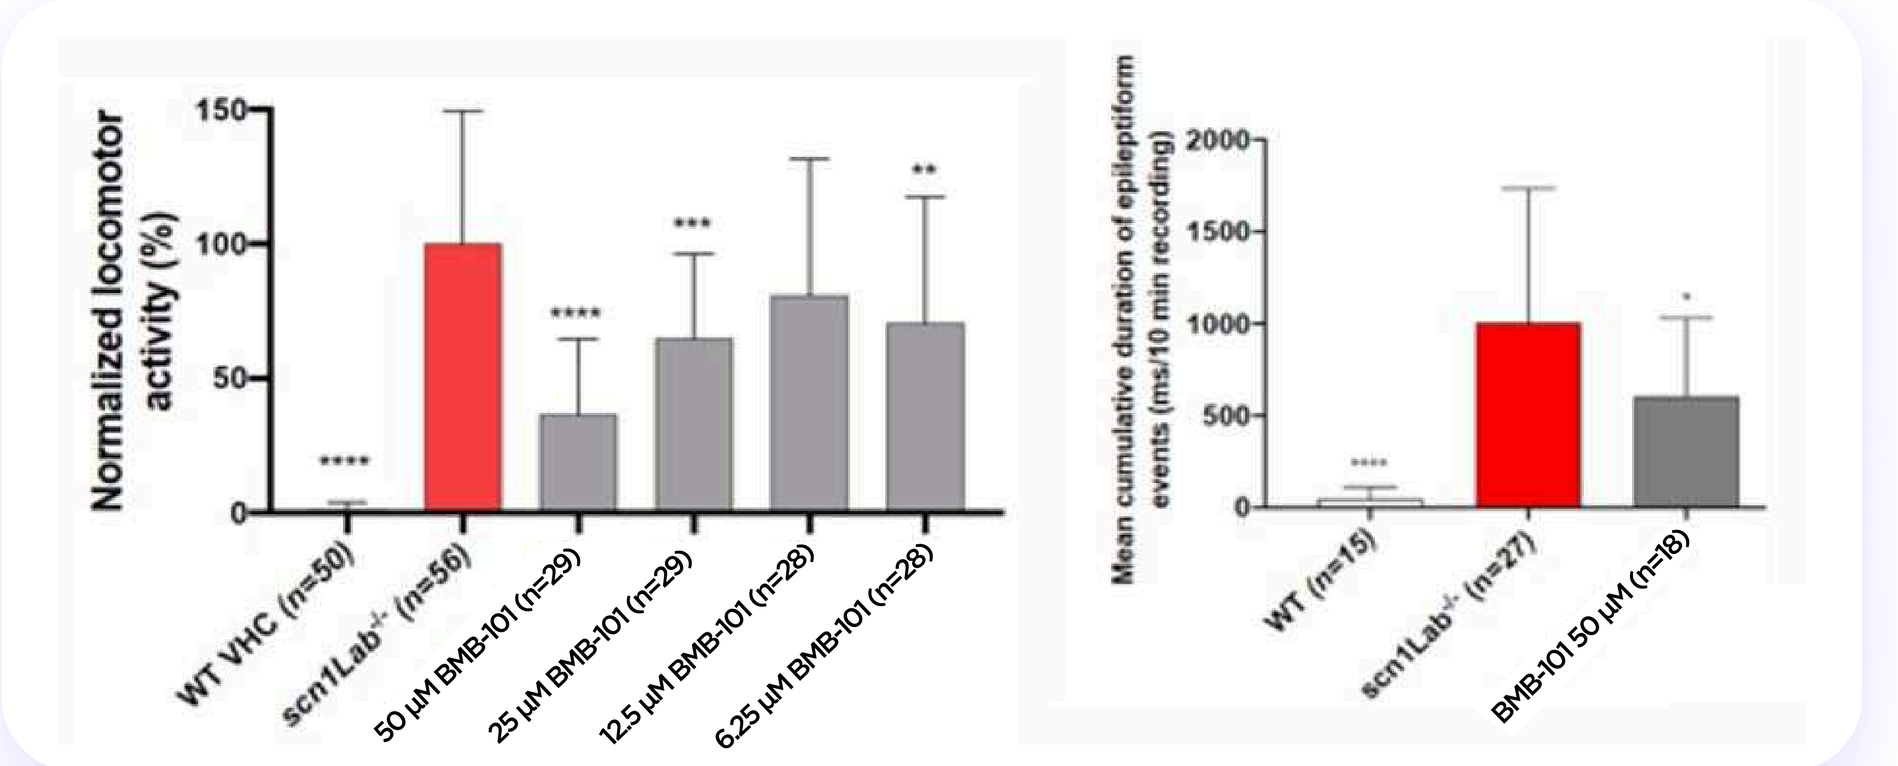
Normalized locomotor activity (%)
WT VHC (n=50)
scn1Lab-/- (n=56)
50 μM BMB-101 (n=29)
25 μM BMB-101 (n=29)
12.5 μM BMB-101 (n=28)
6.25 μM BMB-101 (n=28)
Mean cumulative duration of epileptiform events (ms/10 min recording)
WT (n=15)
scn1Lab-/- (n=27)
BMB-101 50 μM (n=18)

Mouse 6-Hz (44 mA) (psychomotor seizure model)

- Drug-resistant psychomotor seizures were induced 30 minutes to 1 hour after intraperitoneal injection of vehicle (VHC), positive control valproate (VPA), and BMB-101
- In the mouse 6-Hz (44 mA) psychomotor seizure model, BMB-101 reduced the seizure duration at 30 minutes and 1 hour after injection
- Proprietary biased agonists developed by Bright Minds Biosciences (BMB-101 and its analog JJ42) demonstrate efficacy in other preclinical in vivo models of epilepsy and impulsivity-related disorders:
 - Binge eating: ~50% reduction of binge eating episodes, as well as noted weight loss
 - Substance use disorder: ~60% reduction of fentanyl self-administration
 - Maximum electroshock model[5]

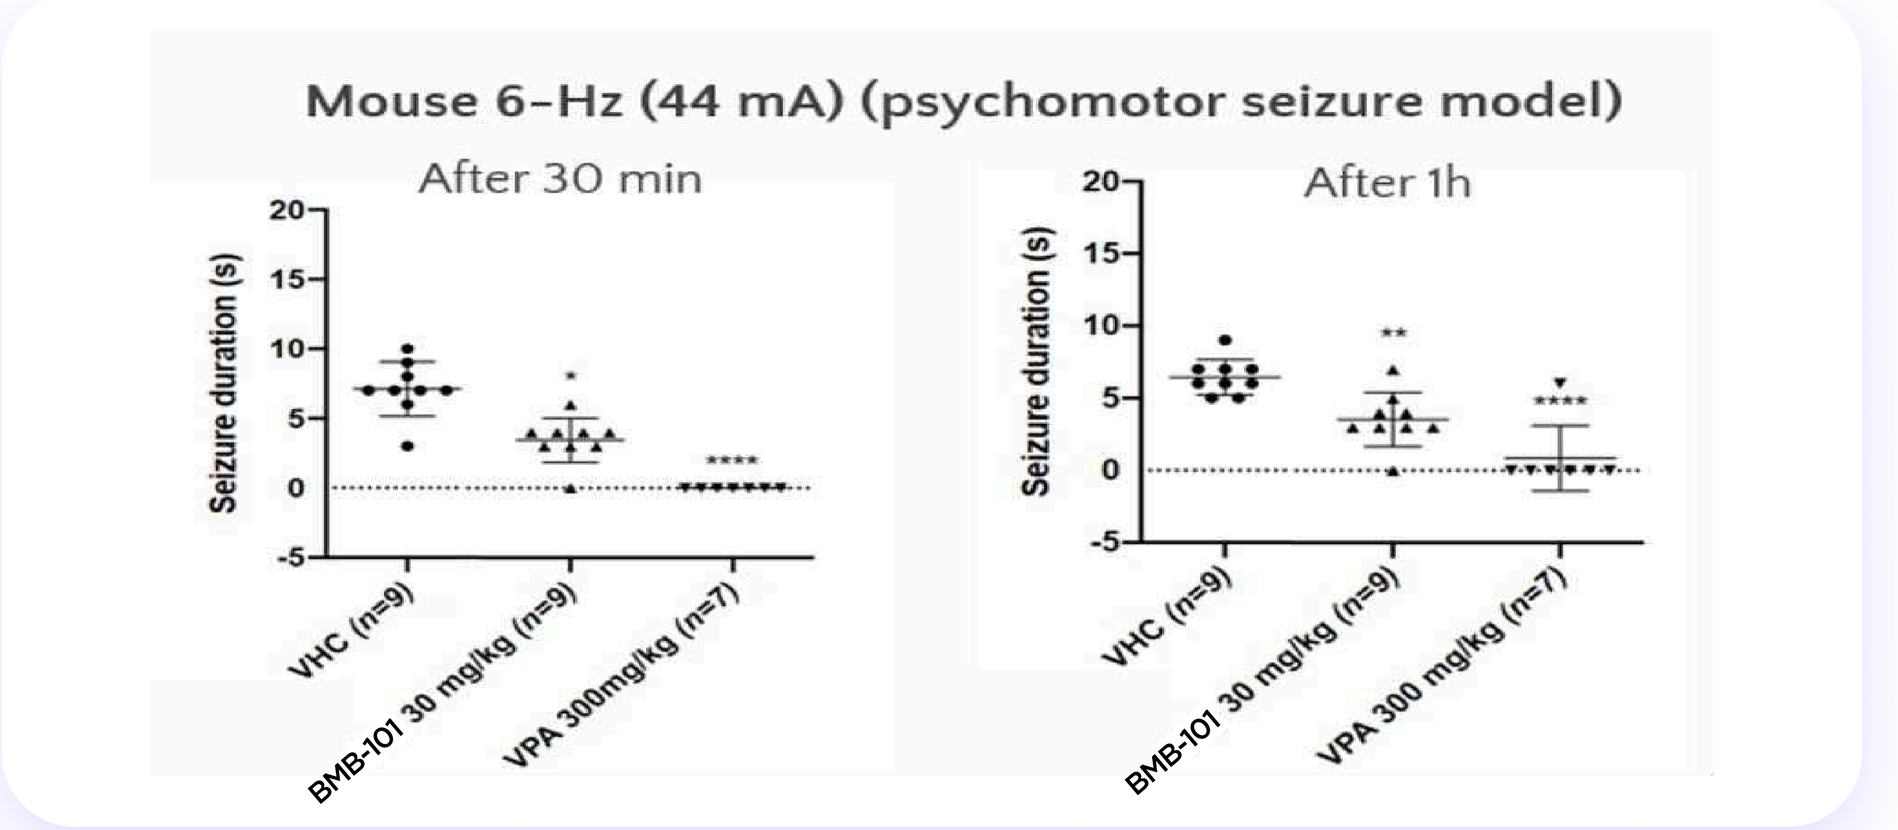
Mouse 6-Hz (44 mA) (psychomotor seizure model)
After 30 min
After 1h
Seizure duration (s)
VHC (n=9)
BMB-101 30 mg/kg (n=9)
VPA 300mg/kg (n=7)

Innovative approach: Biased-agonism

- G-protein biased agonists (or functionally selective agonists) represent a new generation of medications, with potentially improved efficacy and/or less tolerance
- Examples of biased G-protein coupled receptor agonists with improved long-term efficacy include:
 - Tirzepatide: G-protein biased GLP1 agonist for weight management (developed by Eli Lilly)
 - Oliceridine: G-protein biased μ-opioid receptor for severe pain (developed by Trevena)
 - CT-868: G-protein biased GLP1 agonist (developed by Carmot, acquired by Roche)

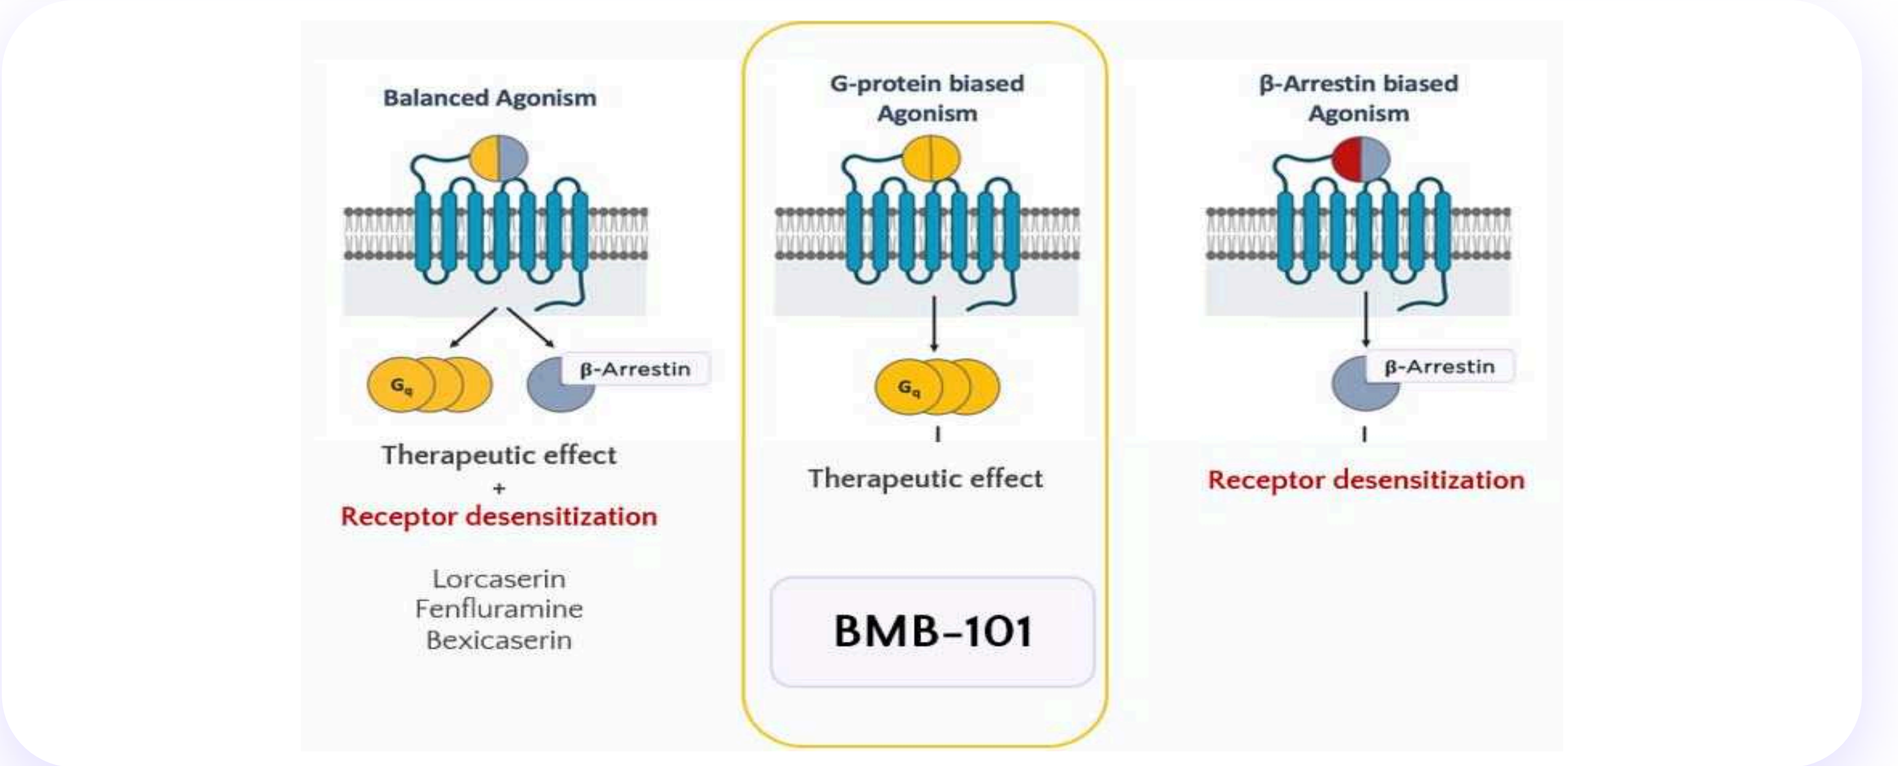
Balanced Agonism
G-protein biased Agonism
β-Arrestin biased Agonism
β-Arrestin
Therapeutic effect
+
Receptor desensitization
Lorcaserin
Fenfluramine
Bexicaserin
Therapeutic effect
BMB-101
Receptor desensitization

Conclusions

- **β-arrestin activation is associated with steric hindrance of further 5-HT$_{2C}$ Gq activation, 5-HT$_{2C}$ receptor internalization and impeding further stimuli of the 5-HT$_{2C}$ receptor. This mechanism has been proposed to cause long-term inefficacy of lorcaserin and fenfluramine.**
- **BMB-101 is the only Gq-biased 5-HT$_{2C}$R selective agonist in clinical development (Phase 2 ongoing), only targeting one of the G-protein coupled receptor pathways without β-arr1 and β-arr2 recruitment, suggesting lack of tolerance in humans**
- **BMB-101 demonstrated anti-seizure activity in preclinical models and is currently in Phase 2 in patients with developmental and epileptic encephalopathy (DEE) and absence epilepsy (BREAKTHROUGH study, NCT06401538)**

References

1. Rankovic et al. Bioorg Med Chem Lett. 2016;26(2):241-50
2. Felsing et al. Eur J Pharmacol. 2019; 5;848:131-9
3. He et al. J Neurosci. 2021;41(26):5734-46
4. Ren et al. J Med Chem. 2025;68(11):10599-618.
5. Vasilkevich et al. Selective 5-HT2C Agonists for the Treatment of Rare Epileptic Disorders. Presented at the Society for Neuroscience, October 5-9, 2024, Chicago